OPEN-LABEL FOLLOW-UP OF THE PEPITES STUDY (PEOPLE) TO EVALUATE THE LONG-TERM EFFICACY AND SAFETY OF EPICUTANEOUS PEANUT IMMUNOTHERAPY IN PEANUT-ALLERGIC CHILDREN

David M. Fleischer,[1] Gordon Sussman,[2] Kirsten Beyer,[3] Jonathan Hourihane,[4] Roxanne C. Oriel,[5] Lara Ford,[6] Wayne Shreffler,[7] Dianne E. Campbell,[6,8] Todd D. Green,[8,9] Robin Mukherjee,[8] Philippe Bégin[10]

[1]Children's Hospital Colorado, University of Colorado, Aurora, CO, USA [2]Gordon Sussman Clinical Research, Toronto, Ontario, Canada [3]Charité Universitätsmedizin Berlin, Berlin, Germany [4]Department of Pediatrics & Child Health, University College Cork, Ireland [5]Icahn School of Medicine at Mount Sinai, Kravis Children's Hospital, Department of Pediatrics, Division of Allergy and Immunology, The Elliot and Roslyn Jaffe Food Allergy Institute, New York, NY, USA [6]Children's Hospital at Westmead, Sydney, Australia [7]Massachusetts General Hospital, Boston, MA, USA [8]DBV Technologies SA, Montrouge, France [9]UPMC Children's Hospital of Pittsburgh and University of Pittsburgh School of Medicine, Pittsburgh, PA, USA [10]CHU Sainte-Justine, Montreal, Quebec, Canada

RATIONALE

- Peanut allergy (PA) is a common and often lifelong food allergy; with limited treatment options, the standard of care is careful avoidance of peanuts and prompt administration of epinephrine in cases of allergic reactions[1]
- As a result of PA, many children and their families experience stress due to fear of accidental exposure, difficulties managing everyday food-related activities, and decreased quality of life[2]
- Epicutaneous immunotherapy (EPIT) for PA aims to utilize the unique immune properties of the skin to induce desensitization in allergic individuals[3]
- An epicutaneous peanut patch containing 250 µg of peanut protein, Viaskin Peanut 250 µg (VP250), has demonstrated safety and efficacy in clinical trials of individuals with PA,[4,5] and is currently under investigation as a potential treatment for PA
- The PEOPLE study is a 5-year, ongoing, open-label extension of the completed Peanut EPIT Efficacy and Safety (PEPITES) study,[6] a phase 3, double-blind, placebo-controlled study that evaluated the safety, tolerability, and efficacy of VP250 in children 4 to 11 years of age with PA

OBJECTIVE

- To assess the efficacy, safety, and tolerability of VP250 therapy after 36 months of treatment (including 12 months of VP250 in PEPITES) in an open-label follow-up study in children with PA

METHODS

Study Design

- PEOPLE is an ongoing 5-year open-label extension of the PEPITES study designed to evaluate the long-term efficacy and safety of VP250 in children (aged 4-11 years) with PA **(Figure 1)**
 - o Subjects in PEPITES were randomized (2:1) to receive a VP250 peanut protein patch or a placebo patch applied daily on the interscapular area
- Subjects who completed the 12-month PEPITES study, including a double-blind placebo-controlled food challenge (DBPCFC) at Month 12, were eligible for enrollment in PEOPLE
- Interim 36-month results from subjects who were randomized to receive VP250 in PEPITES and proceeded to open-label VP250 in PEOPLE are reported here
 - o All subjects in PEOPLE received a daily dose of 250 µg peanut protein per patch and were required to remain on a peanut-free diet
- All subjects who reached an eliciting dose (ED) ≥1000 mg at Month 36 were eligible to continue the study for 2 additional months without treatment and were required to maintain a peanut-free diet (sustained unresponsiveness [SU] assessment). A further DBPCFC was administered at the end of this period (Month 38)

Figure 1. Study Design

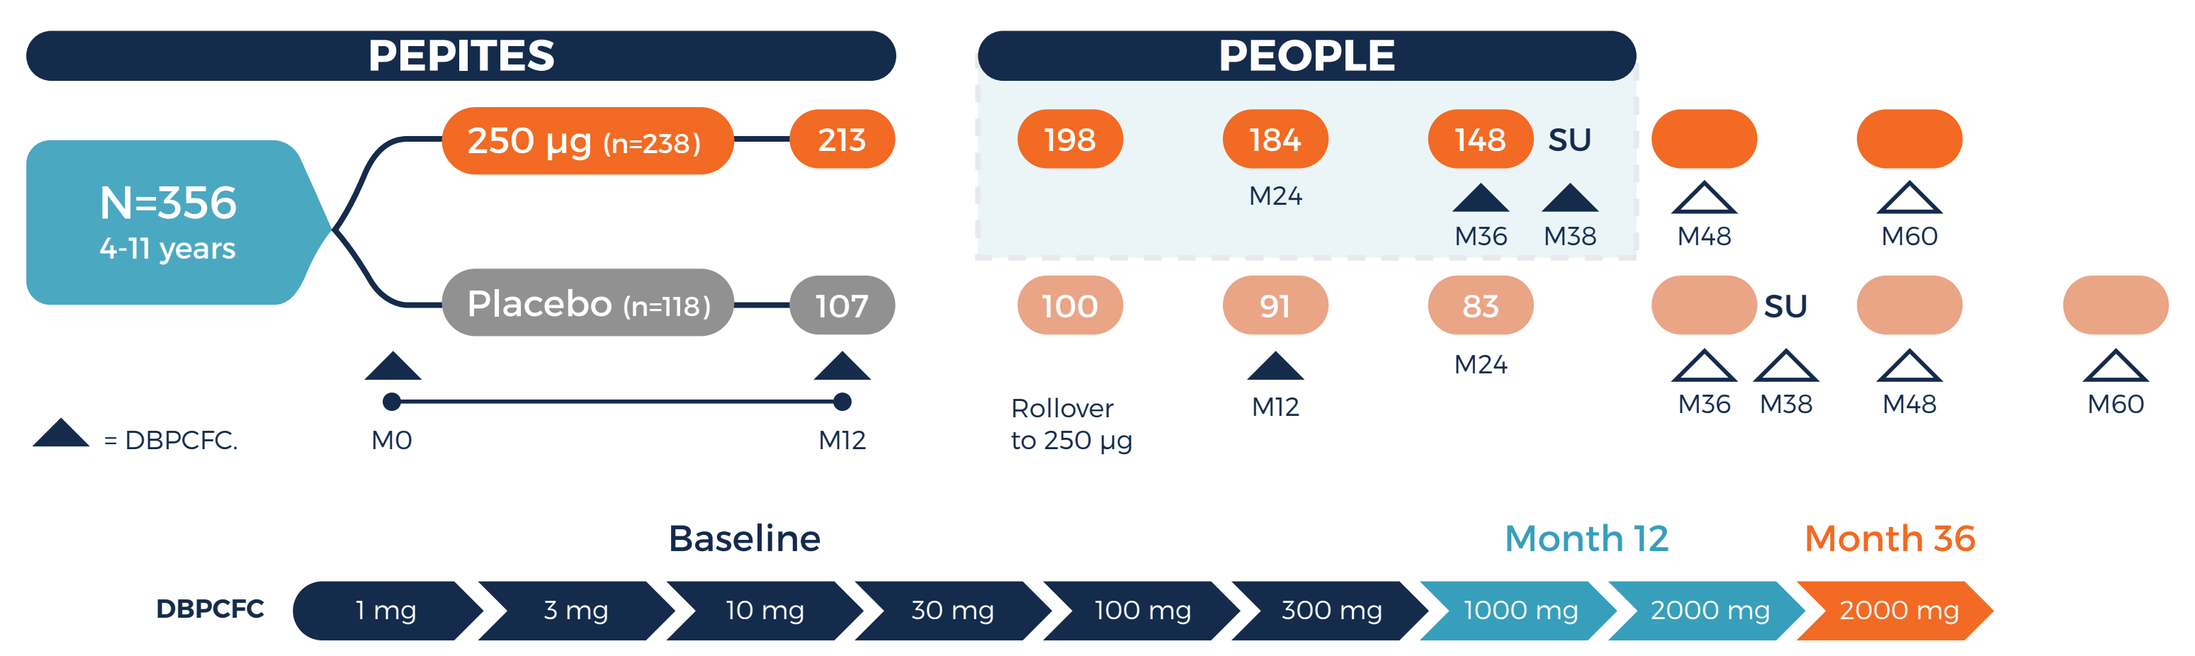
PEPITES
PEOPLE
N=356
4-11 years
250 µg (n=238)
213
Placebo (n=118)
107
198
184
148
SU
100
91
83
M24
M36
M38
M48
M60
M12
Rollover to 250 µg
= DBPCFC.
M0
Baseline
Month 12
Month 36
DBPCFC
1 mg
3 mg
10 mg
30 mg
100 mg
300 mg
1000 mg
2000 mg
2000 mg

Outcomes

- Key outcomes of interest were the percentage of subjects in the VP250+VP250 group who reached an ED of ≥1000 mg after 36 months of active treatment, and the difference between this and the percentage of subjects reaching an ED of ≥1000 mg after 12 months of active treatment
- Additional endpoints included change from baseline to 36 months in ED and in cumulative reactive dose (CRD), safety outcomes, changes in immunoglobulin G4 (IgG4) and immunoglobulin E (IgE) over the 3-year period, and SU assessment

Statistical Analyses

- The safety population consisted of all VP250+VP250 subjects who received at least 1 dose of VP250 in the PEOPLE study
- The completer set was defined as all subjects from the safety set having evaluable DBPCFC results at both Month 12 and Month 36
- The per protocol (PP) set was defined as those subjects who completed all treatment according to the study protocol without major deviations that could affect the assessment of the treatment effect, which included an evaluable DBPCFC performed as required by the protocol at Month 12 and Month 36 and adherence of ≥80%
- ED was determined using the DBPCFC at PEPITES baseline (Month 0) and endpoint (Month 12) and at Month 36 in PEOPLE
 - o At Month 12, the starting dose of the DBPCFC was 1 mg of peanut protein, with escalation up to the highest dose of 2000 mg to reach a maximum total cumulative dose of 3444 mg of peanut protein
 - o At Month 36, the starting dose of the DBPCFC was 1 mg of peanut protein, with escalation up to the highest dose of 2000 mg peanut protein, possibly repeated once to reach a maximum total cumulative dose of 5444 mg of peanut protein
- Categorical variables were summarized using the number of observations and percentages, and continuous variables were summarized using descriptive statistics

RESULTS

Disposition

- Of the 213 subjects who were randomized to the active treatment arm of PEPITES and completed the 12-month trial, 198 (93%) subjects opted to enter PEOPLE **(Figure 2)**

Figure 2. Study Disposition

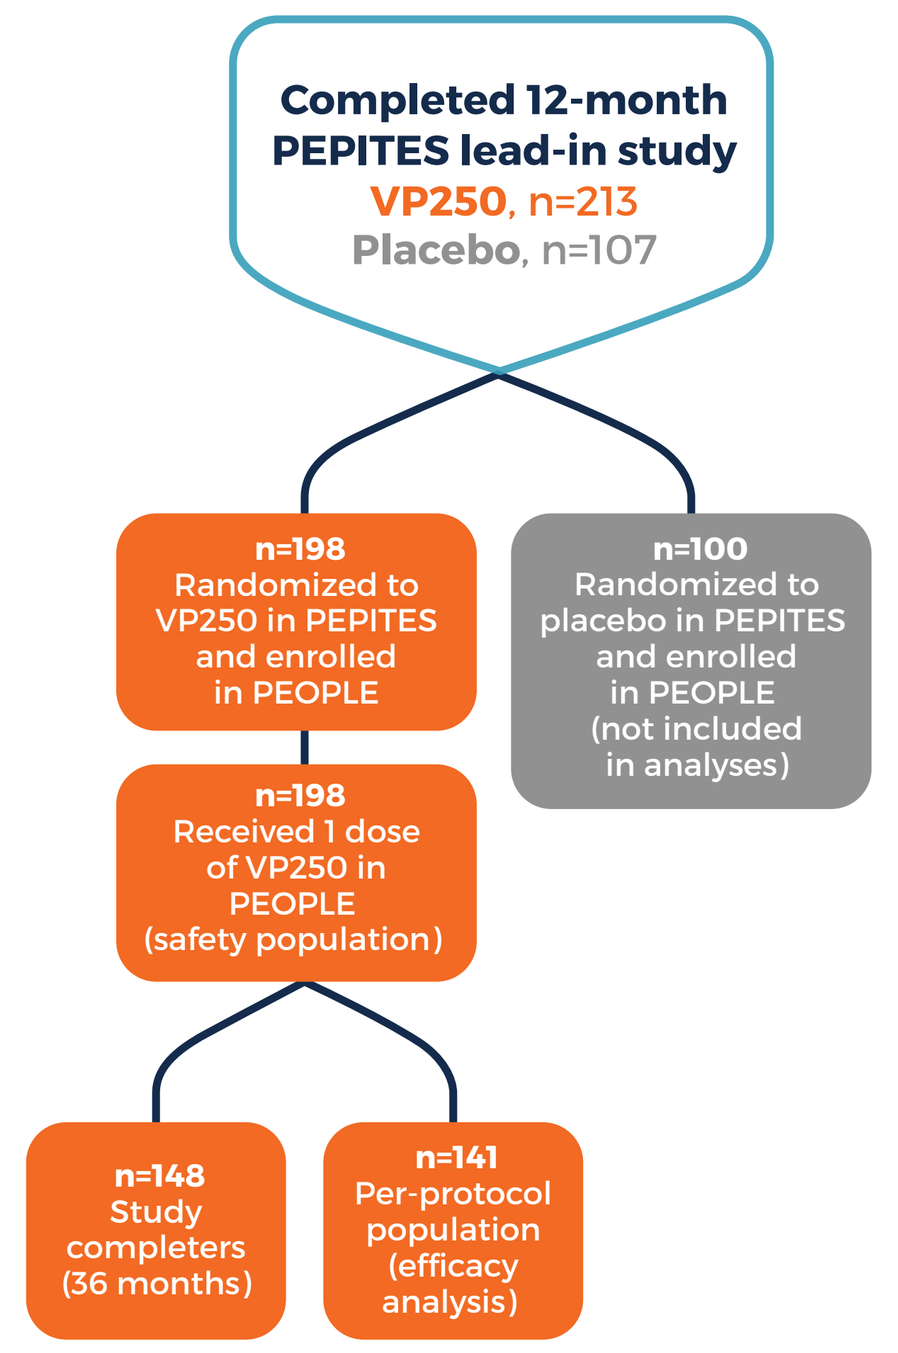
Completed 12-month PEPITES lead-in study
VP250, n=213
Placebo, n=107
n=198 Randomized to VP250 in PEPITES and enrolled in PEOPLE
n=100 Randomized to placebo in PEPITES and enrolled in PEOPLE (not included in analyses)
n=198 Received 1 dose of VP250 in PEOPLE (safety population)
n=148 Study completers (36 months)
n=141 Per-protocol population (efficacy analysis)

EFFICACY

Change in Eliciting Dose

- At Month 36, 51.8% (73/141) of subjects reached an ED of ≥1000 mg, compared with 40.4% (57/141) at Month 12 (11.3% difference; 95% CI: 2.8-19.6) **(Figure 3)**
- Overall, an increase in ED from PEPITES baseline (Month 0) to Month 36 was observed in 75.9% (107/141) of subjects
- Of subjects with baseline ED ≤100 mg, 67.4% (62/92) reached an ED of at least 300 mg
- 13.5% (19/141) of subjects tolerated the full Month 36 DBPCFC of 5444 mg
- In subjects with a baseline (Month 0) ED of ≤10 mg (n=18), there was a 22.5-fold increase in geometric mean ED (95% CI: 10.7-47.3).
- In those with a baseline ED >10 mg (n=123), the geometric mean ED increased 4-fold by Month 36 (95% CI: 3.2-5.0)

Figure 3. Three-Year Changes in Eliciting Dose (PP Population)

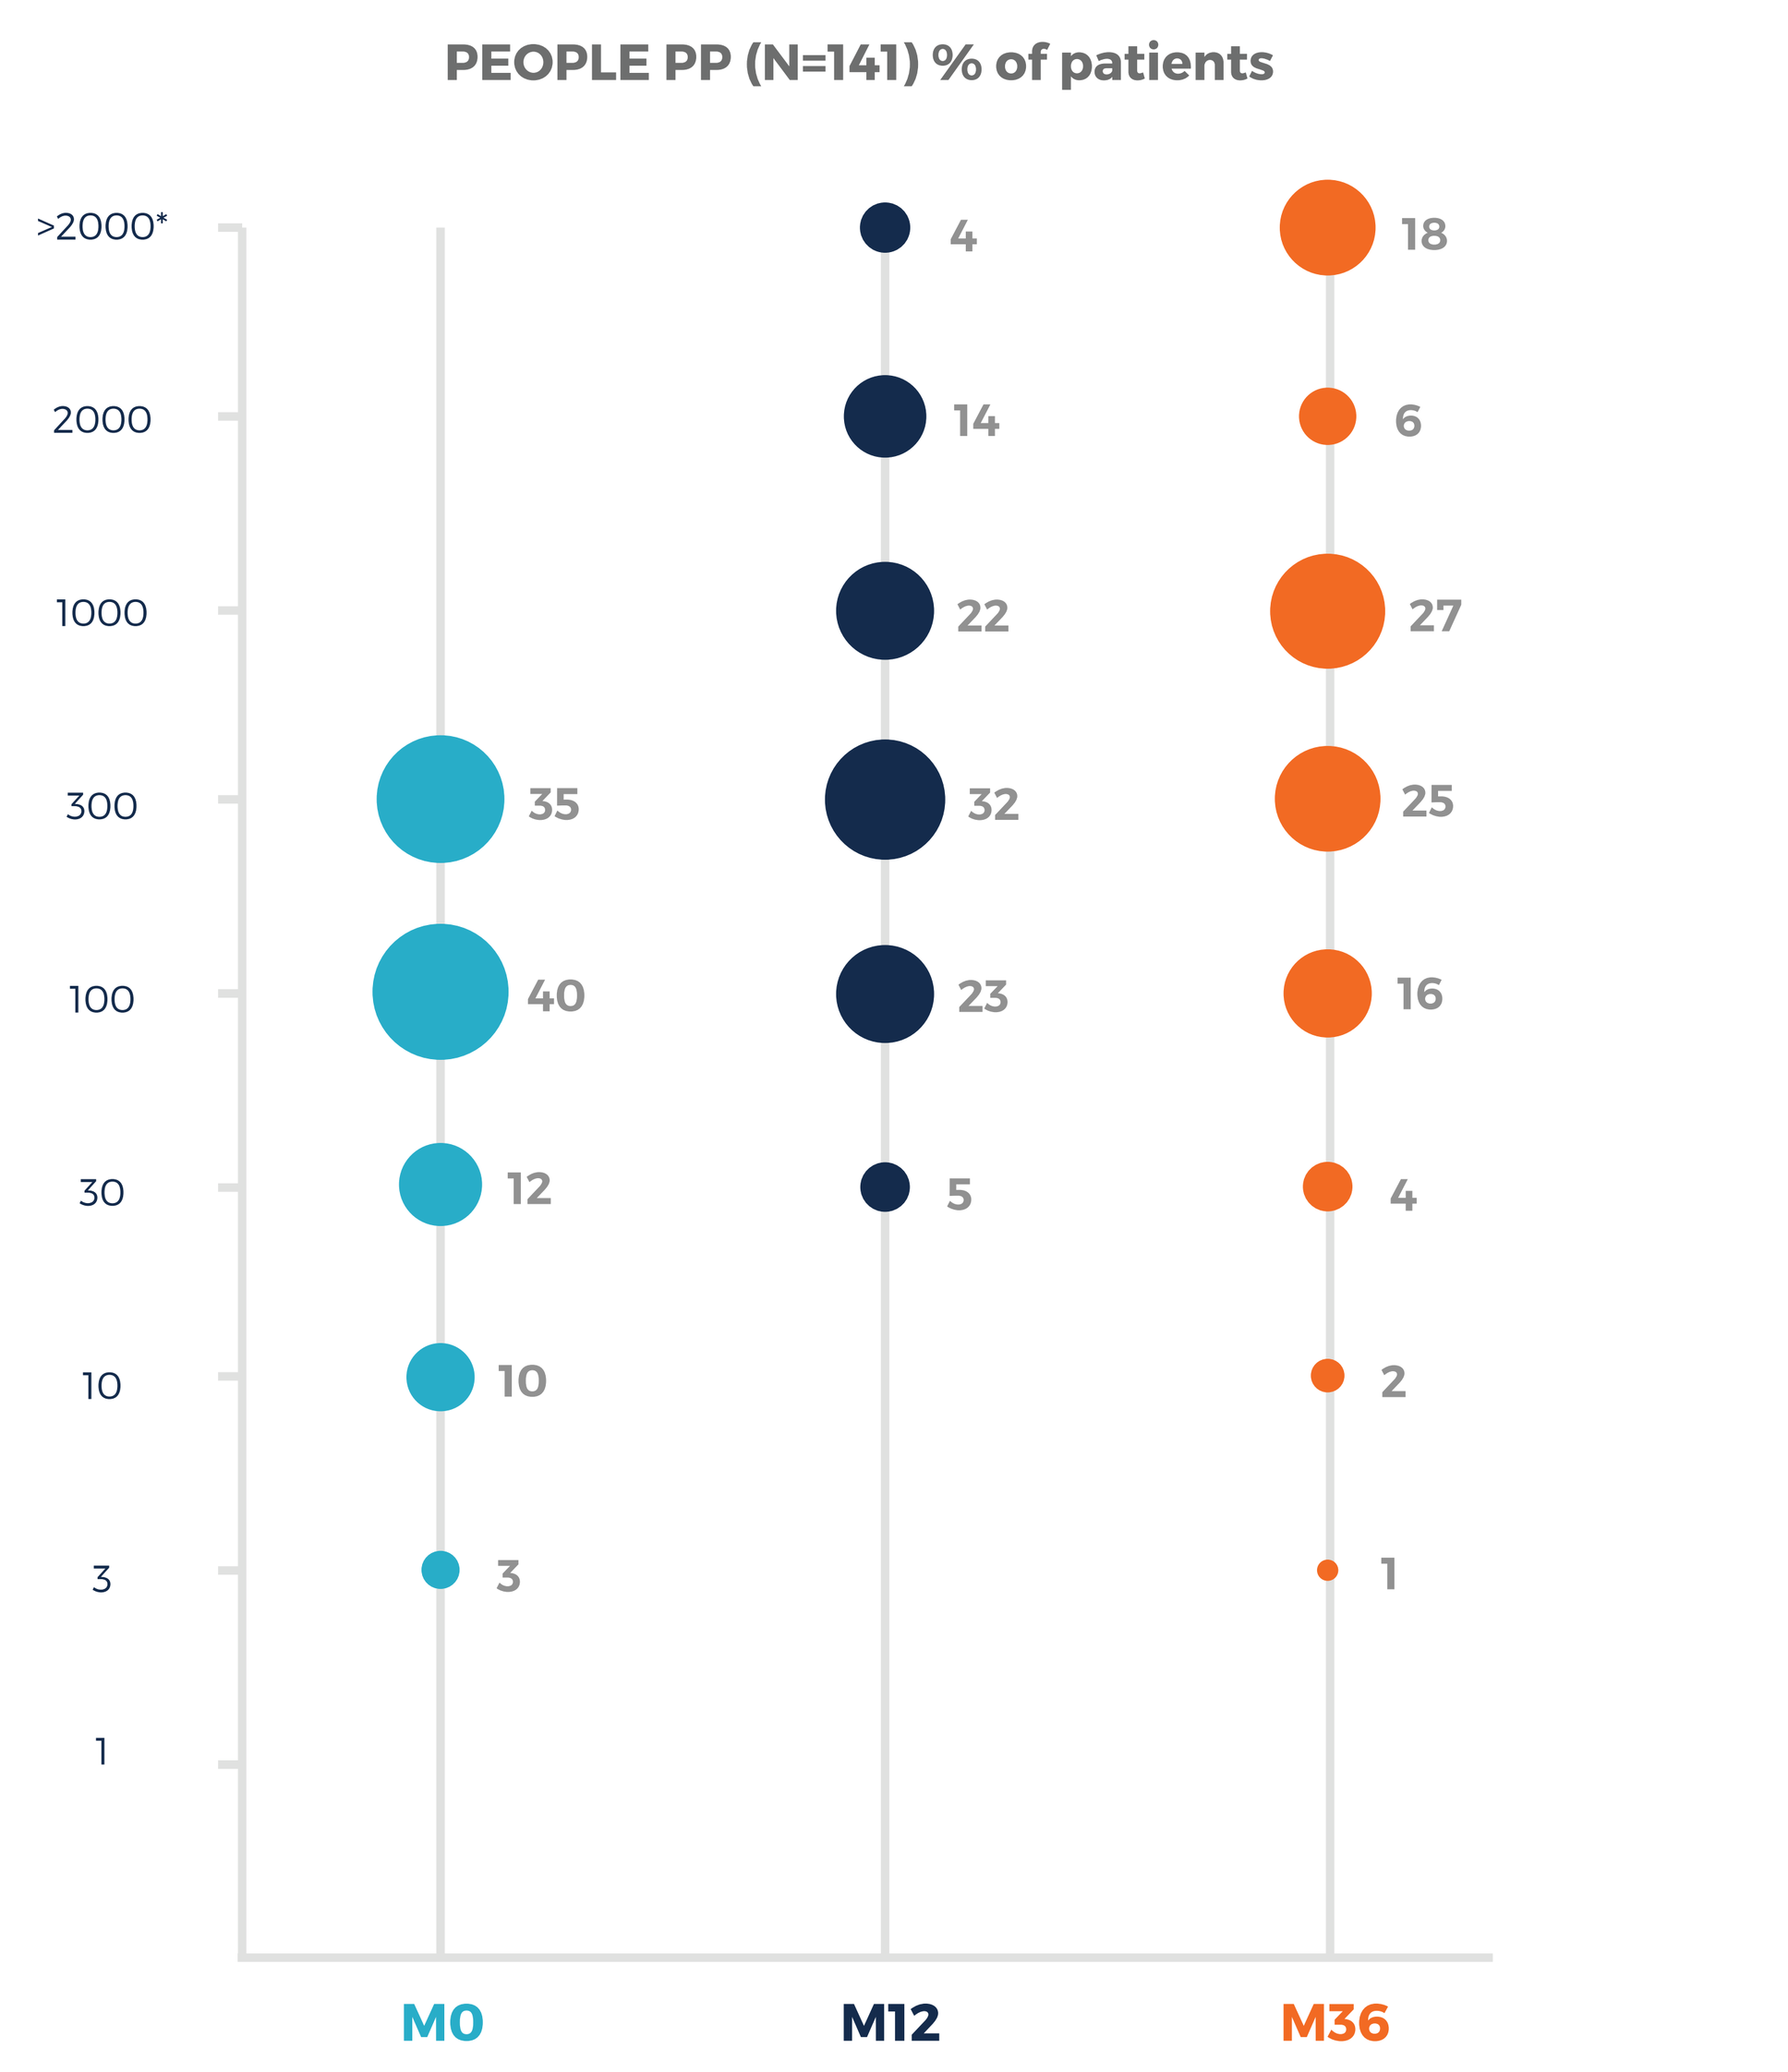
PEOPLE PP (N=141) % of patients
>2000*
2000
1000
300
100
30
10
3
1
35
40
12
10
3
4
14
22
32
23
5
18
6
27
25
16
4
2
1
M0
M12
M36

*ED >2000 mg included subjects: passing the 2000-mg dose at M12 without stopping criteria, passing the 2000-mg dose at M36 without stopping criteria, OR starting the second dose of 2000 mg at M36.

M0, baseline; M12, Month 12; M36, Month 36.

Cumulative Reactive Dose

- At Month 36, the mean CRD was 1768.8 mg, which is roughly equivalent to 6 peanuts **(Figure 4)**
 - o A total of 24.1% (34/141) of subjects reached a CRD of 3444 mg at Month 36 compared with 15.6% (22/141) of subjects at Month 12

Figure 4. Change in Mean CRD From Baseline to Month 36

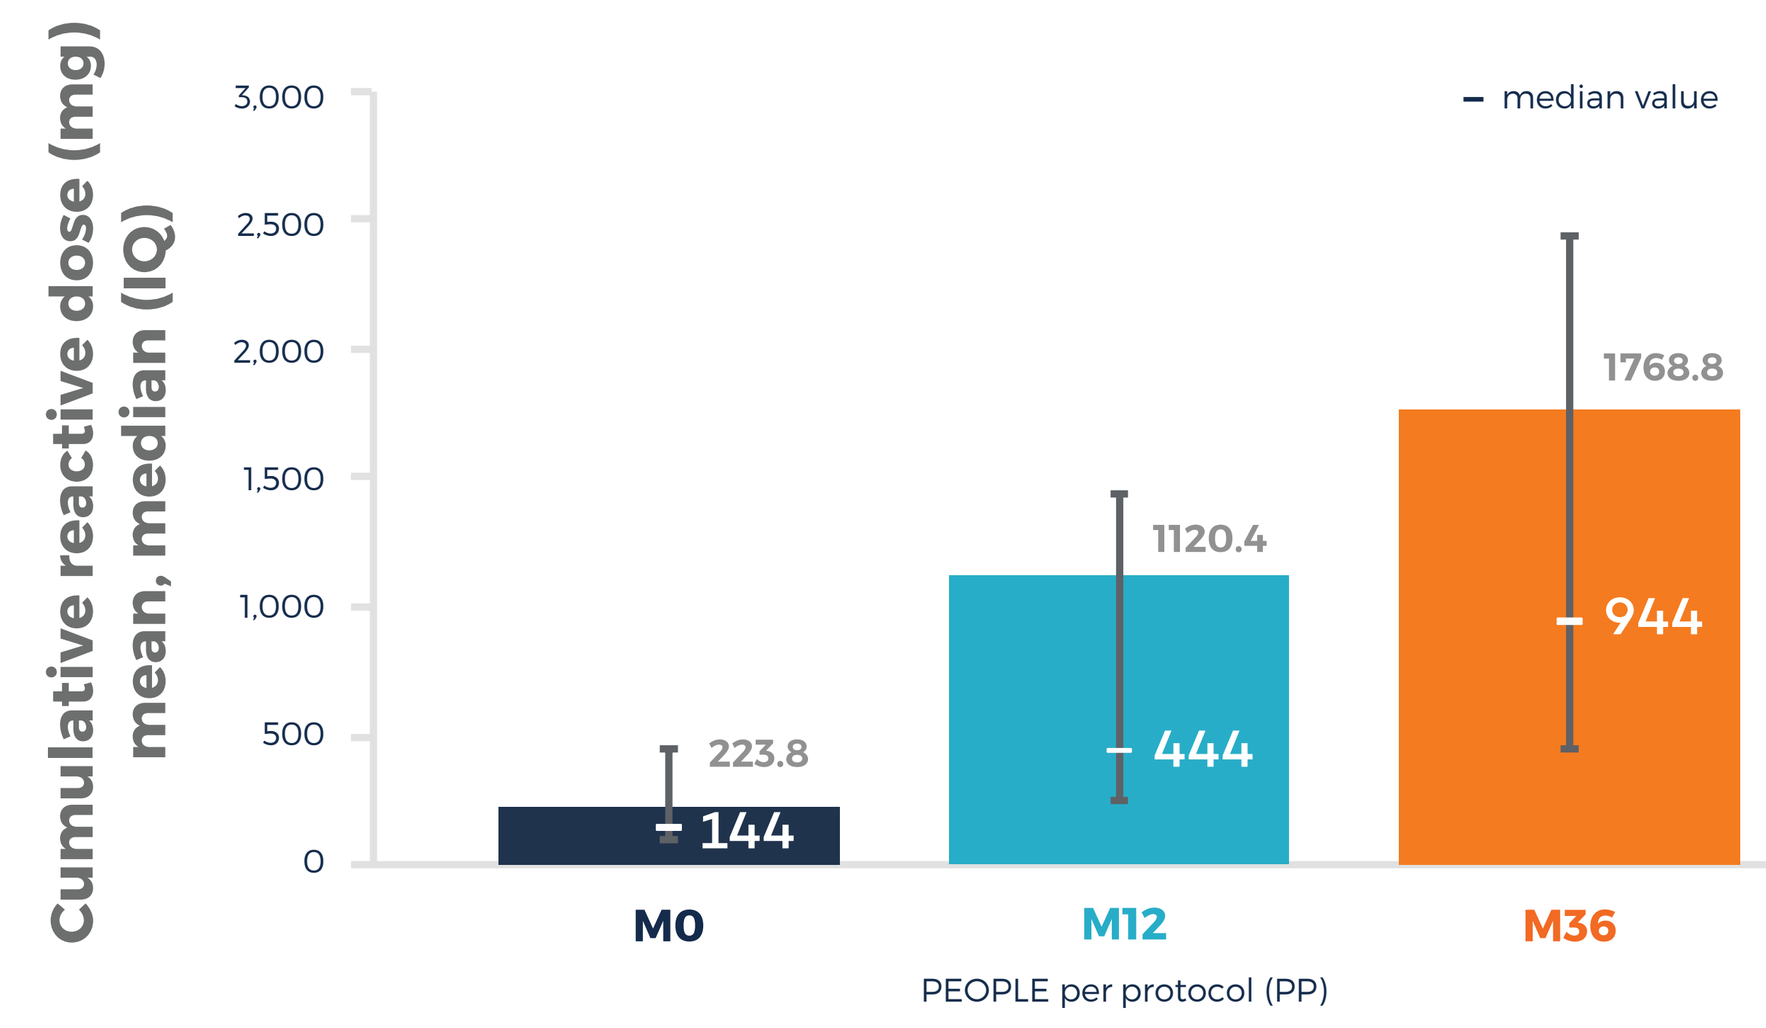
Cumulative reactive dose (mg) mean, median (IQ)
median value
3,000
2,500
2,000
1,500
1,000
500
0
223.8
144
1120.4
444
1768.8
944
M0
M12
M36
PEOPLE per protocol (PP)

Sustained Unresponsiveness

- All 73 subjects who reached an ED ≥1000 mg at Month 36 were eligible to continue the study for 2 additional months without treatment and were required to maintain a peanut-free diet
- Of those, 18 subjects opted to undergo a further DBPCFC at Month 38; 77.8% (14/18) maintained an ED of ≥1000 mg
- Of the 4 subjects who did not maintain an ED of ≥1000 mg at Month 38, 3 subjects reached an ED of 300 mg and 1 subject reached an ED of 100 mg

Change in Peanut IgG4 and IgE Levels

- Peanut IgG4 levels progressively increased throughout the first 18 months of treatment and remained at peak levels for the rest of the treatment to Month 36 **(Figure 5A)**
- IgE levels increased initially at the start of the study and then returned to baseline, remaining around baseline through Month 36 **(Figure 5B)**

Figure 5. Relative Change in Peanut IgG4 (A) and IgE (B) Over the 3-Year Treatment Period in the PP Dataset (n=141)

(A)

(B)

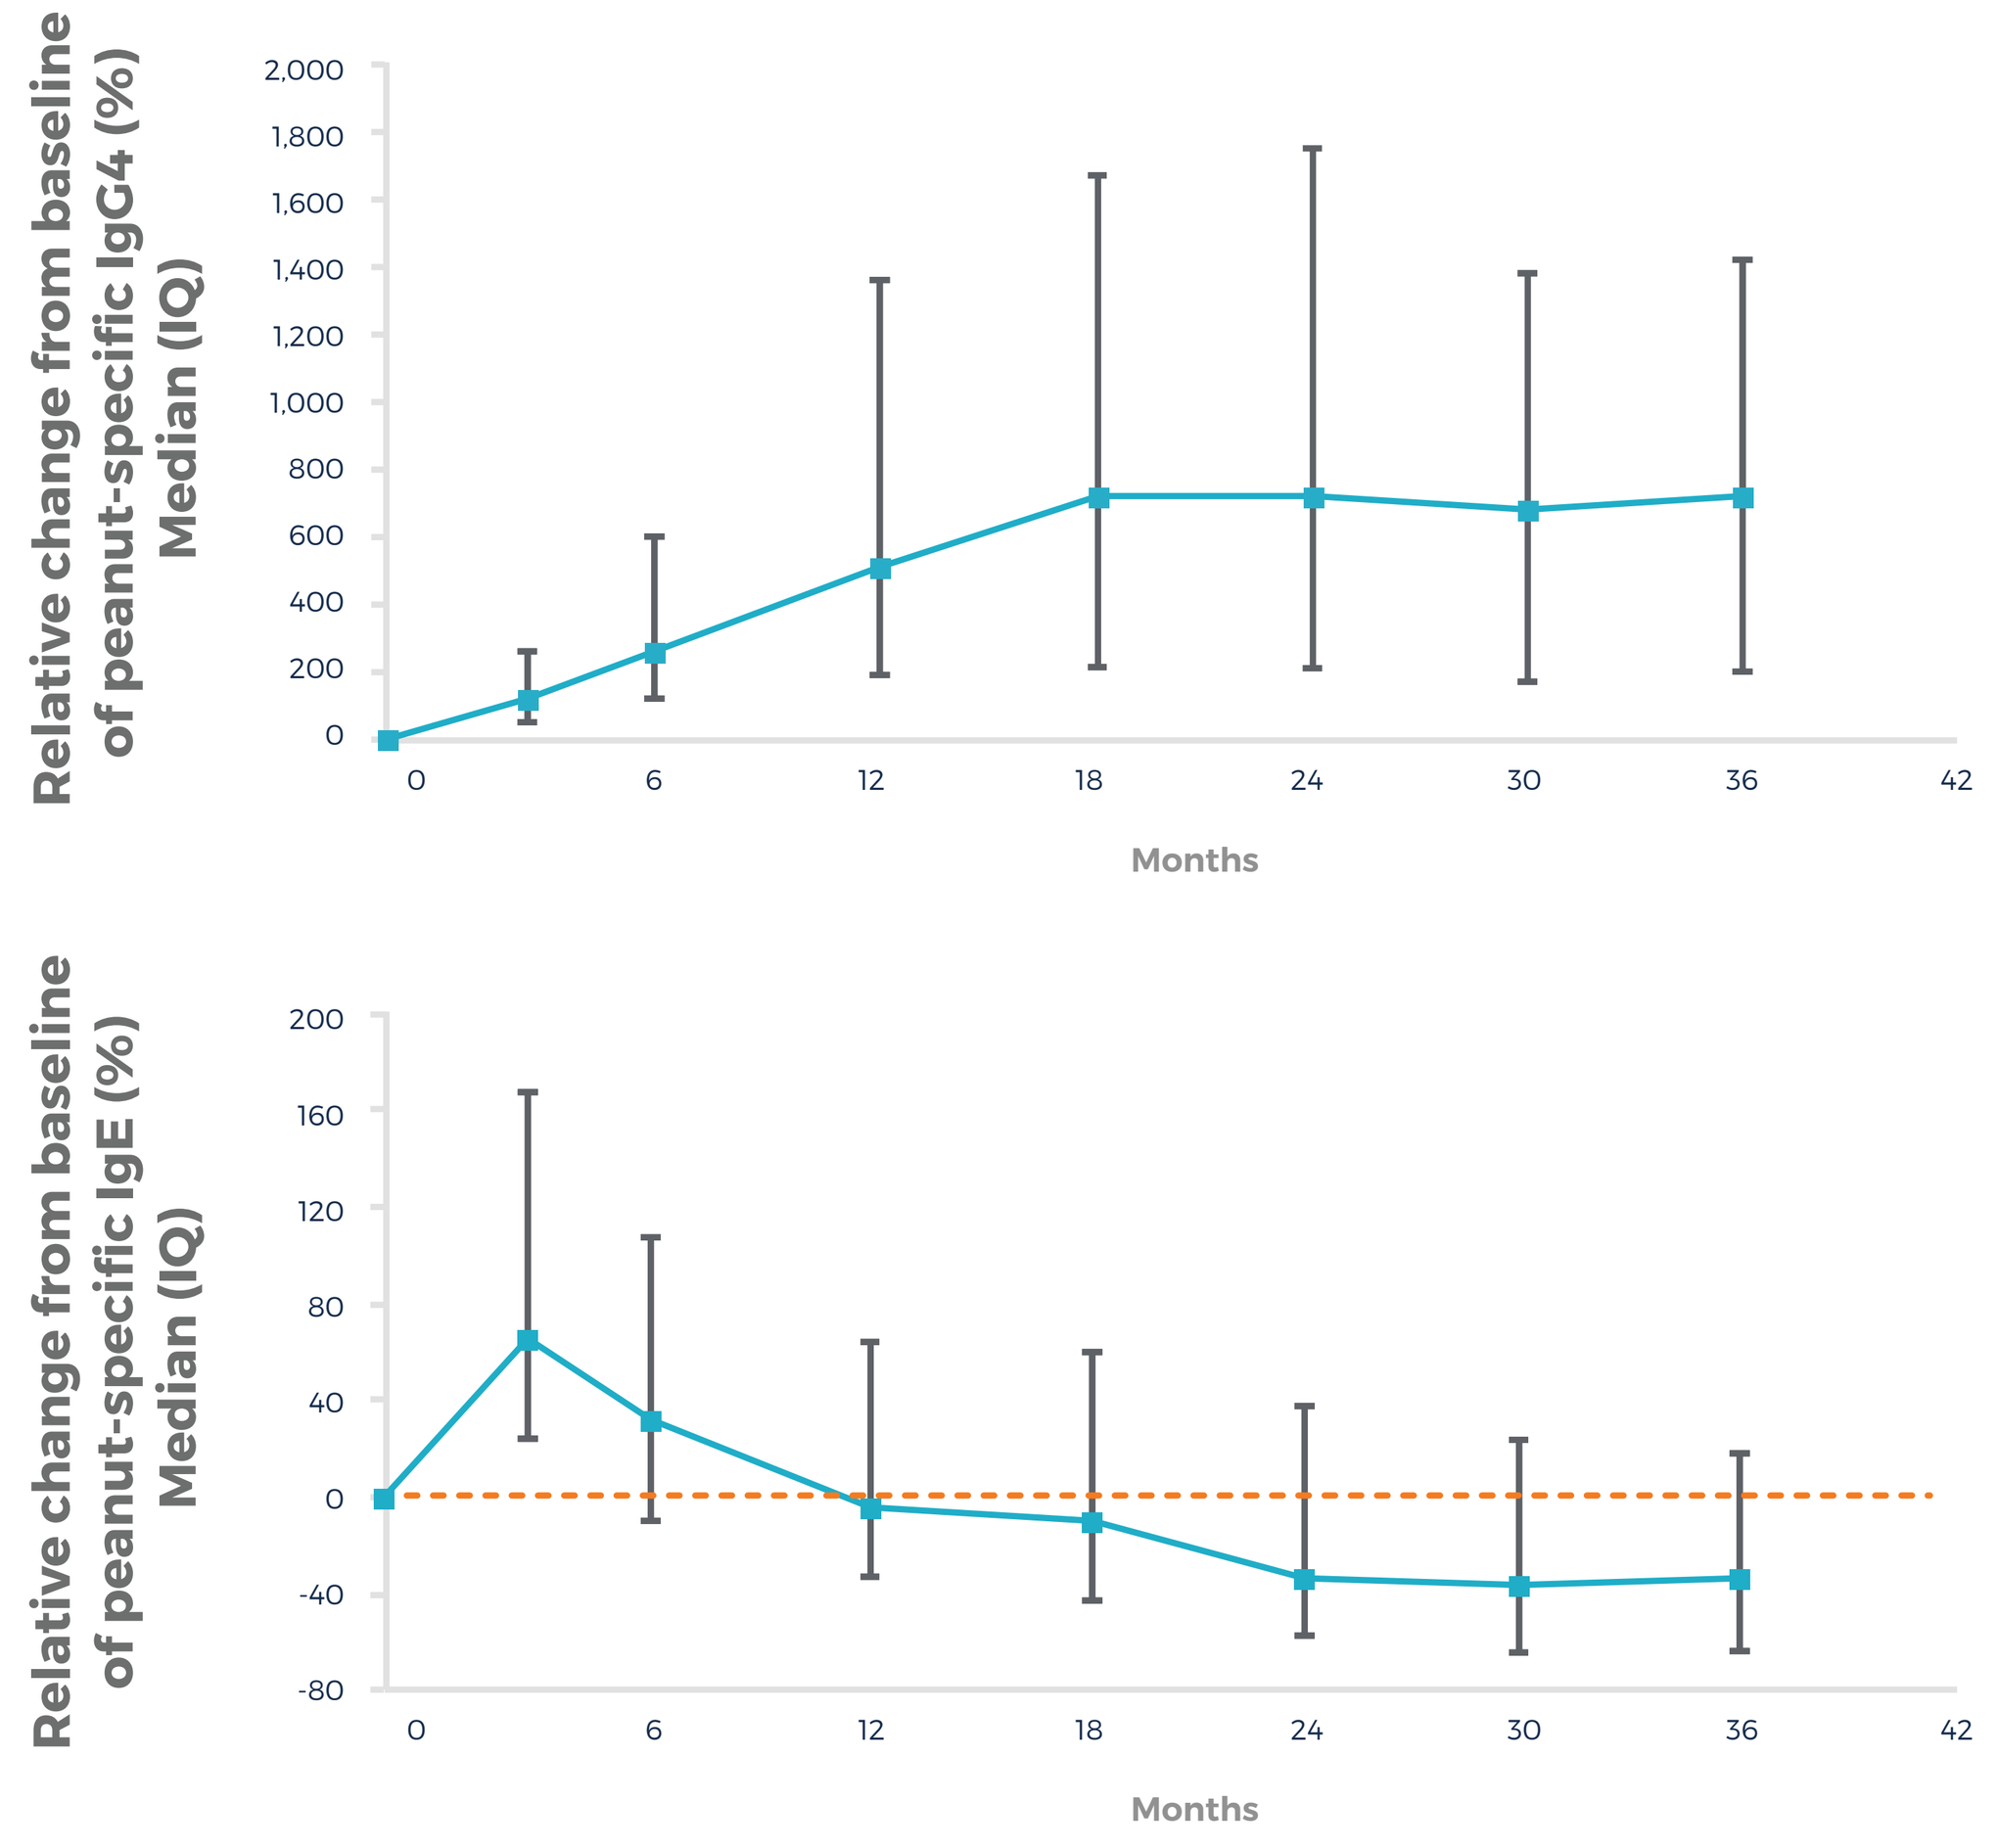
Relative change from baseline of peanut-specific IgG4 (%) Median (IQ)
2,000
1,800
1,600
1,400
1,200
1,000
800
600
400
200
0
Relative change from baseline of peanut-specific IgE (%) Median (IQ)
200
160
120
80
40
0
-40
-80
0
6
12
18
24
30
36
42
Months

SAFETY

Adverse Events

- The incidence of treatment-emergent AEs (TEAEs) was 100% in the safety population (n=198), with discontinuation due to TEAEs reported for 4 (2%) subjects between the start of PEOPLE and Month 36
- The most commonly reported treatment-related TEAEs in PEOPLE were application site reactions, including erythema (63.1%), pruritus (45.5%), and site swelling (20.2%); fewer subjects experienced local skin reactions over time
- In PEOPLE, 1 subject experienced 1 possibly related event of mild anaphylaxis that resolved without treatment; no treatment-related epinephrine intake occurred in PEOPLE

Adherence and Overall Discontinuations

- Over 3 years of treatment, the mean rate of adherence was 98.1% (SD, 4.1) in the safety set
- A total of 50 patients were not part of the completer set due to a not-conducted or non-evaluable DBPCFC at Month 36, including 39 subjects who discontinued the study before Month 36, with 27 discontinuing between Months 30 and 36
 - o DBPCFC fright/distaste was the most common reason (22/50) for completer set exclusion

CONCLUSIONS

- The PEOPLE trial represents the largest long-term trial evaluating peanut immunotherapy to date
- Approximately 3 of 4 subjects experienced an increase in their ED from baseline, which suggests that subjects may have achieved desensitization and a meaningful reduction of risks associated with accidental peanut exposure and demonstrates that a majority of subjects benefited from EPIT for PA with VP250 therapy
- An ED of ≥1000-mg peanut protein was achieved by 51.8% of subjects at Month 36, with a particularly robust response seen in 13.5% of subjects who tolerated a DBPCFC maximum cumulative dose of 5444 mg at Month 36
 - o Additionally, of the subjects who had a baseline ED of ≤100 mg, 67.4% (62/92) reached an ED of at least 300 mg
 - o Median CRD increased approximately 7-fold from baseline (144 mg) to Month 36 (944 mg)
 - o SU was maintained by 78% of subjects who were eligible and opted to participate in the assessment
- VP250 was well tolerated over 3 years of treatment, with a safety profile that was consistent with previous clinical findings
- VP250 may have the potential to durably modify immune response in children with PA
- While longer-term data with EPIT will continue to accumulate with the current study continuing to 5 years, the present study demonstrates that daily EPIT for peanut allergy beyond 1 year leads to continued accrual of benefit, with robust ED improvements that may translate into increased protection from risk of accidental ingestion reactions

REFERENCES

1. Sampson HA, et al. *J Allergy Clin Immunol.* 2014;134(5):1016-1025.e43. **2** Walkner M, et al. *Pediatr Clin North Am.* 2015;62(6):1453-1461. **3.** Bird JA, et al. *World Allergy Organ J.* 2018;11:38. **4.** Jones SM, et al. *Allergy Clin Immunol.* 2016;137(4):1258-1261.e10. **5.** Jones SM, et al. *J Allergy Clin Immunol.* 2017;139(4):1242-1252.e9. **6.** Fleischer DM, et al. *JAMA.* 2019;321(10):946-955.

ACKNOWLEDGMENTS:
This study was sponsored by DBV Technologies. Editorial support in the preparation of this poster was provided by International Meetings & Science (IMsci), funded by DBV Technologies.